4D-C102, a Novel Muscle-Tropic AAV Variant Demonstrates Superior Gene Delivery in Cardiac and Skeletal Muscle Tissues Versus Wild-Type AAV in Human Cells and Non-Human Primates

Melissa Kotterman, Ph.D.[1], Ghezal Beliakoff[1], Julie Nye[1], Roxanne Croze, Ph.D.[1], Christopher Schmitt, Ph.D.[1], Kevin Whittlesey, Ph.D.[1], Gabriel Brooks, M.D.[1], Melissa Quezada[1], Katherine Barglow, Ph.D.[1], David Schaffer, Ph.D.[1,2], Peter Francis, M.D., Ph.D.[1], David Kirn, M.D.[1]

[1] 4D Molecular Therapeutics, Emeryville, CA [2] University of California, Berkeley, CA

Introduction

To unlock the full potential of gene therapy, there is a need to identify novel vectors that show enhanced tropism for target tissues when delivered by clinically relevant routes of administration at commercially feasible doses. Gene replacement strategies leveraging adeno-associated virus (AAV) vectors with high tropism for affected organs may directly address the underlying pathophysiology of neuromuscular diseases, and lysosomal storage diseases, through a combination of cell autonomous correction in target tissues and, when necessary, restoring serum enzyme levels by expression from other key organs, such as heart and skeletal muscle.

Therapeutic Vector Evolution

An industrialized directed evolution approach ("Therapeutic Vector Evolution") was employed in the most relevant animal species (non-human primate; NHP) to discover novel adeno-associated virus (AAV) capsid variants capable of efficient and preferential gene delivery to and robust gene expression in cardiac and skeletal muscle following a single intravenous (IV) administration. The approach resulted in the discovery of 4D-C102, a novel AAV variant capable of efficient gene delivery throughout the primate heart.

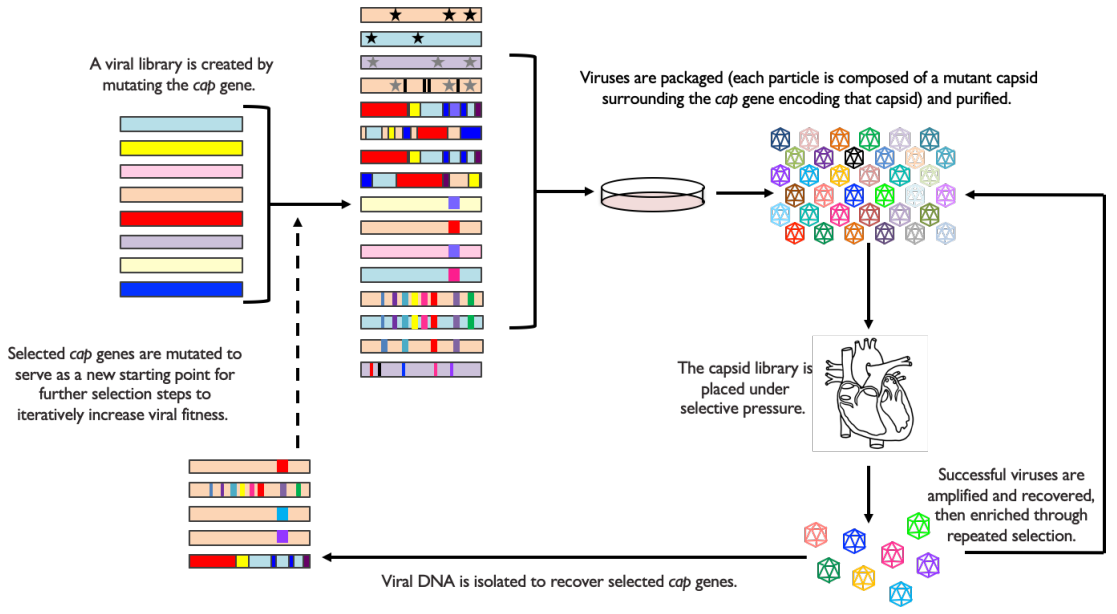
A viral library is created by mutating the cap gene.
Viruses are packaged (each particle is composed of a mutant capsid surrounding the cap gene encoding that capsid) and purified.
Selected cap genes are mutated to serve as a new starting point for further selection steps to iteratively increase viral fitness.
The capsid library is placed under selective pressure.
Successful viruses are amplified and recovered, then enriched through repeated selection.
Viral DNA is isolated to recover selected cap genes.

Figure 1: ***Schematic of Therapeutic Vector Evolution.*** *A viral capsid library comprising 37 proprietary combinations of DNA mutation techniques and cap genes is created. Viruses are then packaged such that each particle is composed of a mutant capsid surrounding the cap gene encoding that capsid and purified. Enrichment of successful clones through repeated selection using in vivo primate delivery to iteratively increase viral fitness. Variants identified as hits during Vector Selection are manufactured as recombinant AAV vectors and characterized for the level of transduction of target cell types and tissues. Adapted from (Kotterman et al. 2014)*

Methods

AAV Transduction of Human Muscle Cells

Human pluripotent stem cell-derived cardiomyocytes (ventricular phenotype) and human primary skeletal muscle cells were transduced with 4D-C102 or control AAV serotypes. Human pluripotent stem cell line ESI-017 was differentiated to cardiomyocytes by small molecule modulation of Wnt signaling for 14 days followed by enrichment for cardiomyocytes by glucose deprivation (Lian et al. 2012; Lian et al. 2013). After 24 days of differentiation, mature cardiomyocytes were used for transduction studies. AAV1, AAV8, AAV9, or 4D-C102.CAG-EGFP were added at increasing multiplicities of infection (MOIs) and incubated for 48 hours. Cells were analyzed six days post-infection. Human skeletal muscle-derived cells (Cook Myosite) were expanded in Myosite Expansion Medium plus Myotonic growth supplement and antibiotics, then differentiated using Myosite Differentiation Medium. After 30 days of differentiation, cells were transduced with AAV8, AAV9, or 4D-C102.CAG-EGFP at increasing MOIs and incubated for 48 hours. Cells were analyzed seven days post-infection.

AAV Administration to Non-Human Primates

Cynomolgus macaques were injected with 1×10^{13} vg/kg AAV8, AAV9, or 4D-C102.CAG-EGFP via the saphenous vein. Approximately seven weeks later, tissues were collected and processed. Tissue samples were analyzed by qPCR analysis for viral genome delivery, immunofluorescence for protein expression, and hemotoxylin and eosin staining for inflammation, respectively. NHPs received daily immunosuppression (30 mg/kg cyclosporin A, 70 mg/kg mycophenolate mofetil) from seven days prior to dosing through study termination. Serum samples were collected for troponin, CK and CKMB analysis.

Robust Gene Delivery & Protein Expression in NHP

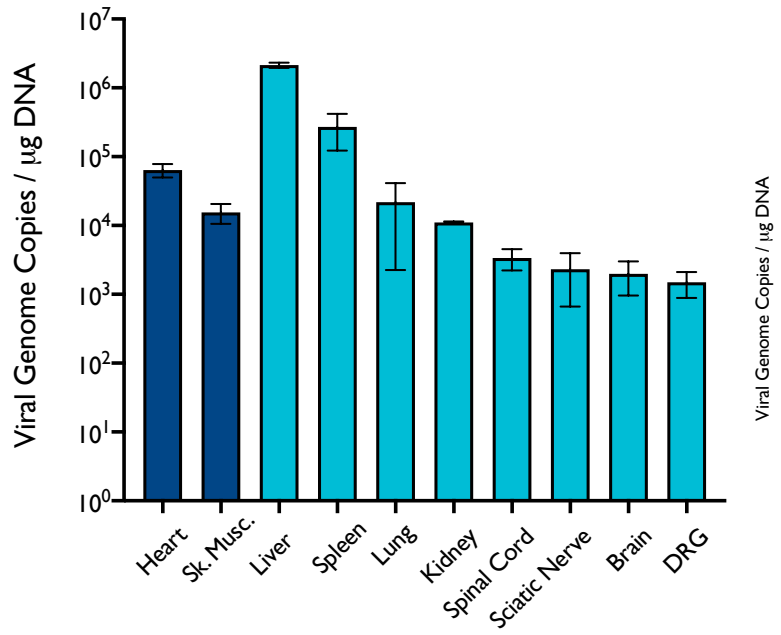
Viral Genome Copies / µg DNA
Heart
Sk. Musc.
Liver
Spleen
Lung
Kidney
Spinal Cord
Sciatic Nerve
Brain
DRG

Figure 2: ***4D-C102 Genome Biodistribution In Vivo in Cynomolgus Macaques****. Quantification of viral genomes in the heart, skeletal muscle, and additional systemic organs by qPCR using primers and probe against the EGFP transgene. Viral genomes were detected in all heart samples (n = 10 samples per NHP) and skeletal muscle samples (n = 27 samples per NHP). Mean ± standard error; n = 3 NHP (n = 5 samples per liver, n = 1 samples per spleen, n = 1 samples per lung, n = 2 total samples for kidney, n = 3 samples per spinal cord, n = 1 samples per sciatic nerve, n = 7 samples per brain, n = 3 samples per DRG). Dark blue bars indicate tissues of primary interest.*

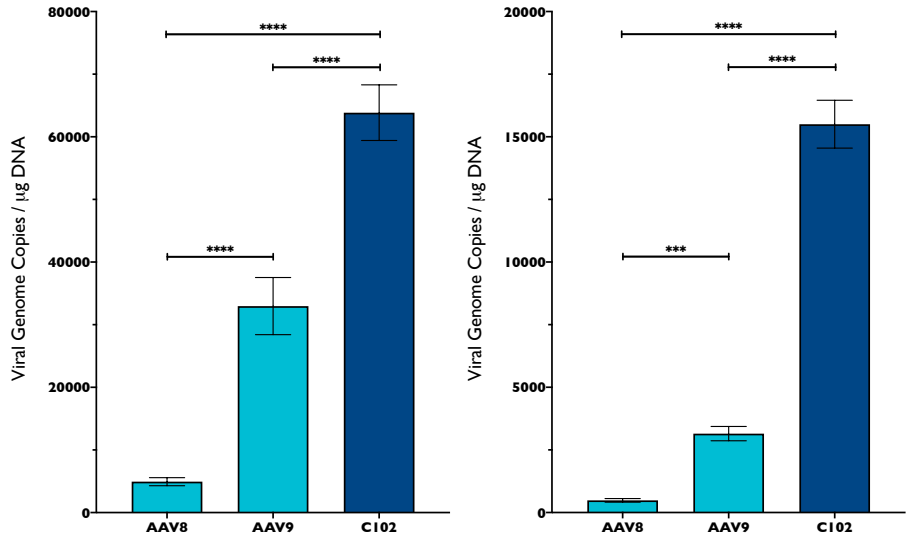
Viral Genome Copies / µg DNA
AAV8
AAV9
C102

Figure 3: ***4D-C102 Genome Delivery In Vivo in Cynomolgus Macaques.*** *Quantification of viral genomes in the heart (left) and skeletal muscle (right) by qPCR using primers and probe against the EGFP transgene following delivery of 1x10¹³ vg/kg of AAV8, AAV9, or 4D-C102.CAG-EGFP. Mean ± standard error; n = 3 NHP per group. n = 10 heart samples per NHP, n = 27 skeletal muscle samples per NHP. *** = p < 0.0005, **** = p < 0.0001, ANOVA*

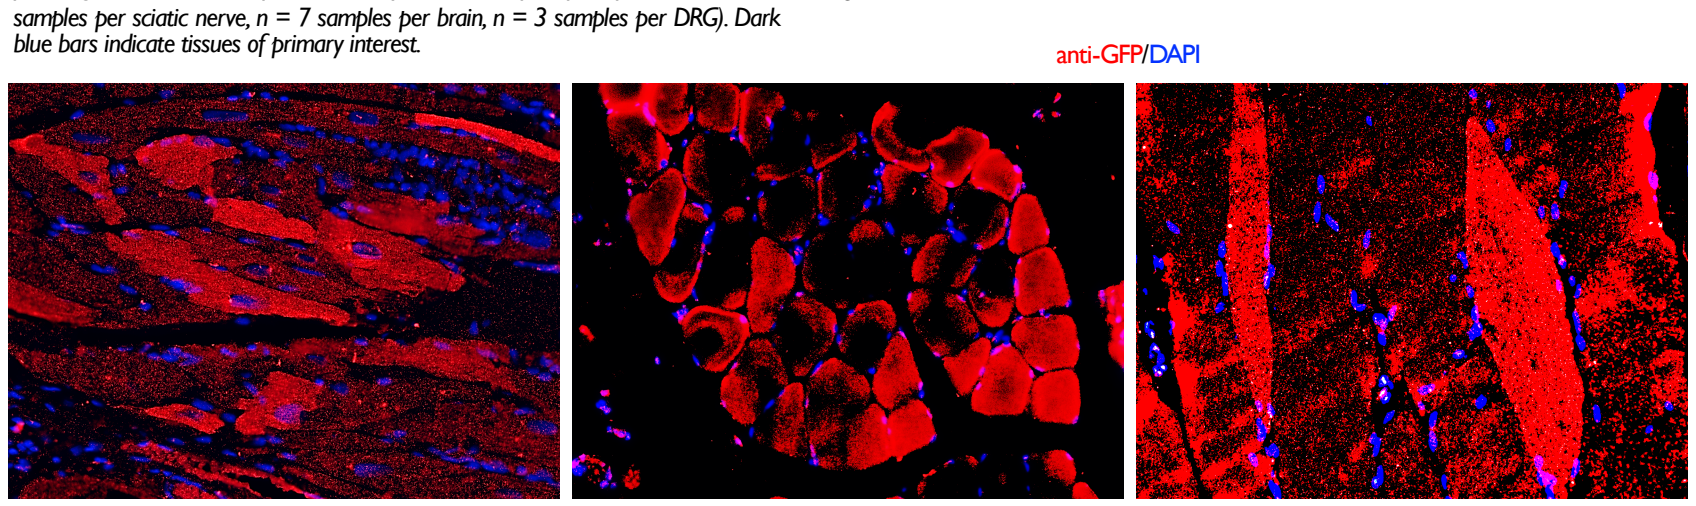
anti-GFP/DAPI

Figure 4: ***4D-C102 Protein Expression In Vivo in Cynomolgus Macaques.*** *GFP expression in the left ventricle (left), distal triceps (middle), and distal vastus lateralis (right) following delivery of 4D-C102.CAG-EGFP on Day 50 ± 2 post-IV injection of 1x10¹³ vg/kg.*

No Histopathological or Clinical Pathology in NHP

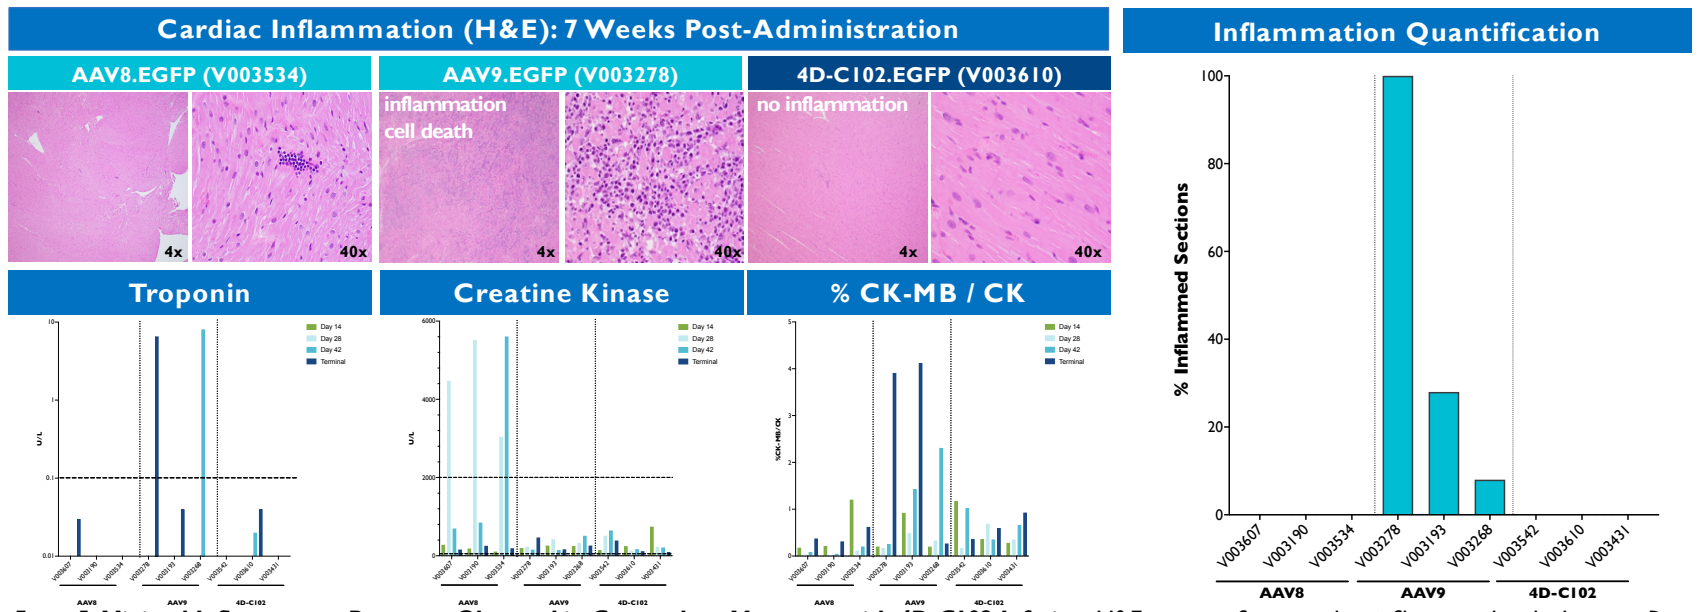
Cardiac Inflammation (H&E): 7 Weeks Post-Administration
AAV8.EGFP (V003534)
AAV9.EGFP (V003278)
4D-C102.EGFP (V003610)
inflammation
cell death
no inflammation
Troponin
Creatine Kinase
% CK-MB / CK
Inflammation Quantification
% Inflammed Sections
AAV8
AAV9
4D-C102

Figure 5: ***Minimal Inflammatory Response Observed in Cynomolgus Macaques with 4D-C102 Infusion.*** *H&E staining of mononuclear infiltrates within the heart on Day 50 ± 2 post-IV injection of 1x10¹³ vg/kg of AAV8, AAV9, or 4D-C102.CAG-EGFP (illustrative micrographs shown; top left). Percentage of cardiac sections containing >25% inflamed area by H&E (minimum of 20 cardiac sections per NHP; right). Quantification of the level of troponin, creatine kinase (CK), and percent creatine kinase muscle/brain (CK-MB) of total CK present in serum sampled at 4 timepoints post-IV injection (bottom left).*

Efficient Transduction of Human Muscle Cells

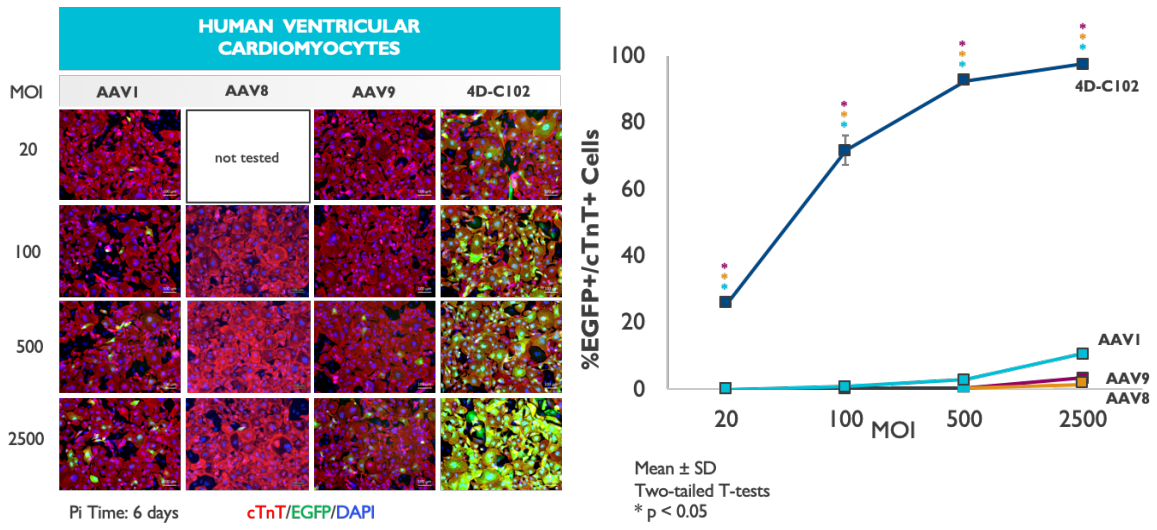
HUMAN VENTRICULAR CARDIOMYOCYTES
MOI
AAV1
AAV8
AAV9
4D-C102
not tested
Pi Time: 6 days
cTnT/EGFP/DAPI
%EGFP+/cTnT+ Cells
MOI
Mean ± SD
Two-tailed T-tests
* p < 0.05

Figure 6: ***Human Cardiomyocytes Transduction is Superior with 4D-C102 versus AAV1, AAV8, AAV9*** *(left) Representative immunofluorescence images, (right) flow cytometry quantification % EGFP+ human stem cell-derived ventricular cardiomyocytes 6 days post-infection. Mean ± standard deviation, * = p < 0.051, two-tailed Student's T-test n = 3 wells.*

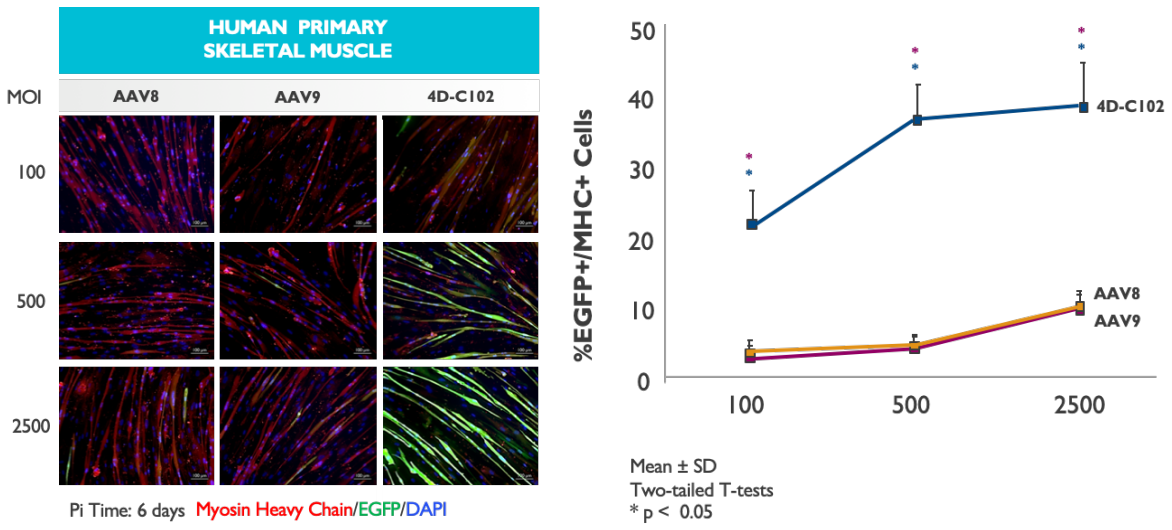
HUMAN PRIMARY SKELETAL MUSCLE
MOI
AAV8
AAV9
4D-C102
Pi Time: 6 days
Myosin Heavy Chain/EGFP/DAPI
%EGFP+/MHC+ Cells
Mean ± SD
Two-tailed T-tests
* p < 0.05

Figure 7: ***Human Cardiomyocytes Transduction is Superior with 4D-C102 versus AAV1, AAV8, AAV9*** *(left) Representative immunofluorescence images, (right) flow cytometry quantification % EGFP+ human skeletal muscle-derived cells 7 days post-infection. Mean ± standard deviation, * = p < 0.05, two-tailed Student's T-test n = 3 wells.*

Conclusion

- 4D-C102 represents first use of directed evolution in NHP to identify a vector engineered for multiple muscle tissue transduction by IV administration.
- 4D-C102 demonstrated superior gene delivery in human cardiomyocytes and skeletal muscle cells *in vitro*.
- 4D-C102 demonstrated superior gene delivery in NHP *in vivo* following IV administration.
- The tropism profile of 4D-C102 represents a significant advance over existing AAV serotypes.
- 4D-C102 is well-suited for the development of gene therapies for neuromuscular conditions, lysosomal storage diseases involving muscle tissue, and muscular dystrophies.
- 4DMT is currently developing 4D-310 as a next generation gene therapeutic for the treatment of Fabry disease (Abstracts #140 & #594)

References

- Kotterman M, Schaffer D (2014). Engineering adeno-associated viruses for clinical gene therapy. Nat. Rev. Genet. 15(7):445-51
- Lian X, Hsiao C, Wilson G, Zhu K, Hazeltine LB, Azarin SM, Raval KK, Zhang J, Kamp TJ, Palecek SP (2012). Robust cardiomyocyte differentiation from human pluripotent stem cells via temporal modulation of canonical Wnt signaling. Proc Natl Acad Sci. 109(27):E1848-57.
- Lian X, Zhang J, Azarin SM, Zhu K, Hazeltine LB, Bao X, Hsiao C, Kamp TJ, Palecek SP. (2013). Directed cardiomyocyte differentiation from human pluripotent stem cells by modulating Wnt/β-catenin signaling under fully defined conditions. Nat Protoc. 8(1):162-75.